A Phase III Trial of Muparfostat (PI-88) as Adjuvant Therapy in Patients with Hepatitis Virus Related Hepatocellular Carcinoma (HV-HCC) after Resection

P.-J.Chen[1], P.-H. Lee[2], K.-H. Han[3], J. Fan[4], T.T. Cheung[5], R-H. Hu[6], S.W. Paik[7], W.-C. Lee[8], G.-Y. Chau[9], L.-B. Jeng[10], HJ. Wang[11], J.Y. Choi[12], C.-L Chen[13], M. Cho[14], M.-C. Ho[15], C.-C. Wu[16], KS. Lee[17], Y. Mao[18], F.-C. Hu[19], K.-L. Lai[20]

[1]Graduate Institute of Clinical Medicine and Department of Medical Research, National Taiwan University and National Taiwan University Hospital, Taipei, TW, [2]Department of Surgery, National Taiwan University Hospital, Taipei, TW, [3]Department of Internal Medicine, Yonsei University College of Medicine, Seoul, KR, [4]Liver Cancer Institute, Zhongshan Hospital, Cancer Center, Institute of Biomedical Sciences, Fudan University, Shanghai, CN, [5]Department of Surgery, Queen Mary Hospital and University of Hong Kong, Hong Kong, HK, [6]Department of Surgery, College of Medicine, National Taiwan University Hospital, Taipei, TW, [7]Department of Gastroenterology, Samsung Medical Center, College of Medicine, Sungkyunkwan University, Seoul, KR, [8]Division of Liver and Transplantation Surgery, Department of General Surgery, Chang Gung Memorial Hospital Linkou Branch, College of Medicine, Chang Gung University, Taipei, TW, [9]Department of General Surgery, Taipei Veterans General Hospital, Taipei, TW, [10]Organ Transplantation Center, China Medical University Hospital, Taichung, TW, [11]Department of Surgery, College of Medicine, Ajou University, Seoul, KR, [12]Division of Hepatology, Department of Internal Medicine, Seoul St. Mary's Hospital, College of Medicine, The Catholic University of Korea, Seoul, KR, [13]Liver Transplantation Center and Department of Surgery, Chang Gung Memorial Hospital Kaohsiung Branch, College of Medicine, Chang Gung University, Kaohsiung, TW, [14]Department of Gastroenterology, Pusan National University Yangsan Hospital, Pusan, KR, [15]Department of Surgery, National Taiwan University Hospital, College of Medicine, National Taiwan University, Taipei, TW, [16]Department of General Surgery, Taichung Veterans General Hospital, Taichung, TW, [17]Department of Gastroenterology, Gangnam Severance Hospital, Seoul, KR, [18]Department of Liver Surgery, Peking Union Medical College Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College, Beijing, CN, [19]Graduate Institute of Clinical Medicine, College of Medicine, National Taiwan University, Taipei, TW, [20]New Drug Development, Medigen Biotechnology Corporation, Taipei, TW

Background

Muparfostat is a mixture of oligosaccharides mimicking heparan sulfate that antagonizes angiogenic growth factors (VEGF, FGF-1, and FGF-2) (Fig.1) and blocks heparanase from cleaving heparan sulfate in the extracellular matrix (ECM) (Fig.2), leading to inhibition of tumor growth and prevention of tumor cell spreading. [1,2] Previous phase II study for HCC patients after curative tumor resection has shown muparfostat (PI-88) to be well tolerated. Administering muparfostat at 160 mg/day is safe and has demonstrated substantial prolongation of disease free survival (DFS) for HCC patients after resection.[3,4] Therefore, we conducted this phase III trial to evaluate the efficacy of muparfostat as adjuvant therapy after HCC resection.

Fig.1 in vitro experiment demonstrates that muparfostat (PI-88) inhibits human placental blood vessel growth.

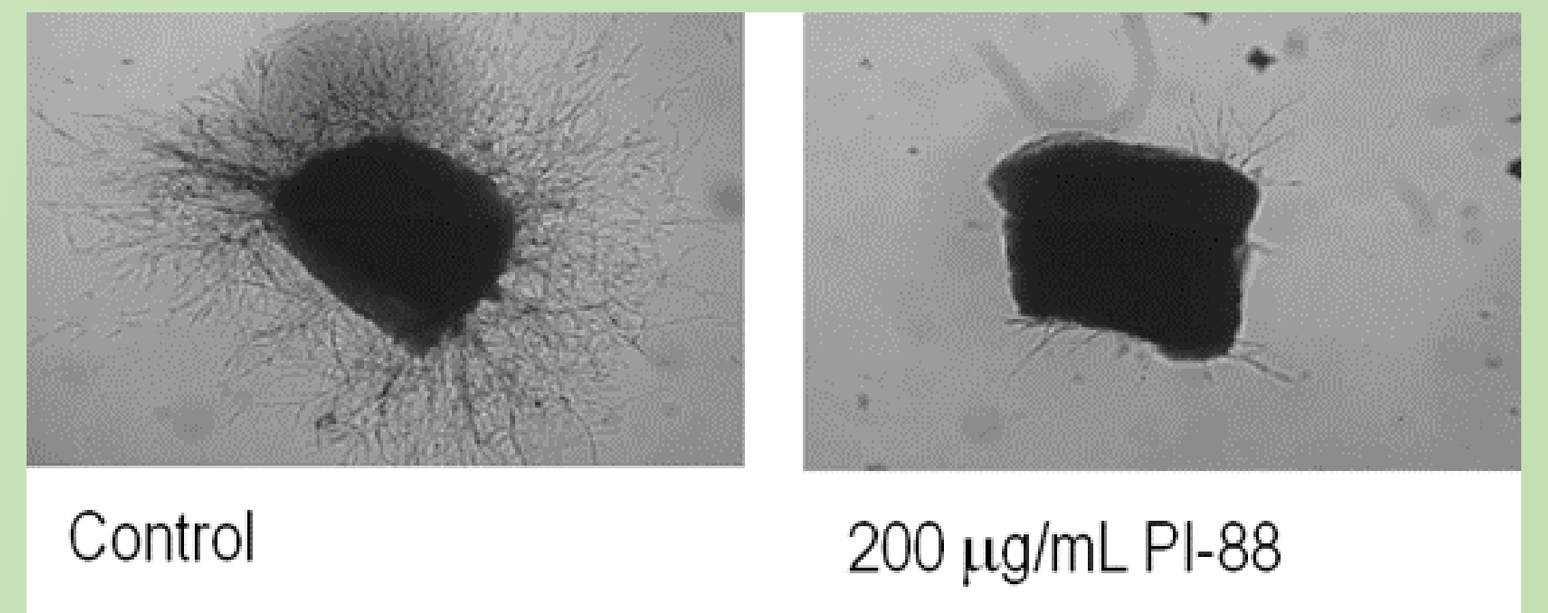

Control
200 μg/mL PI-88

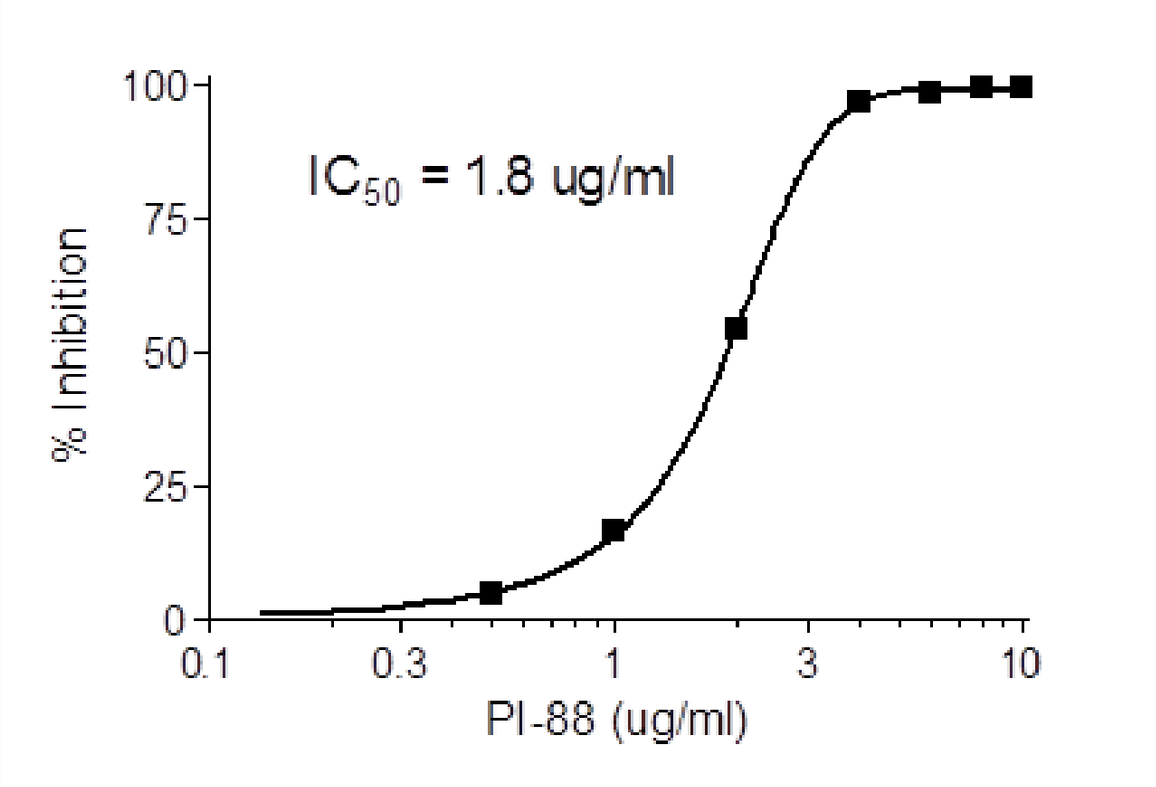

IC50 = 1.8 ug/ml
% Inhibition
PI-88 (ug/ml)

Fig.2 Inhibition of human platelet heparanase by muparfostat (PI-88).

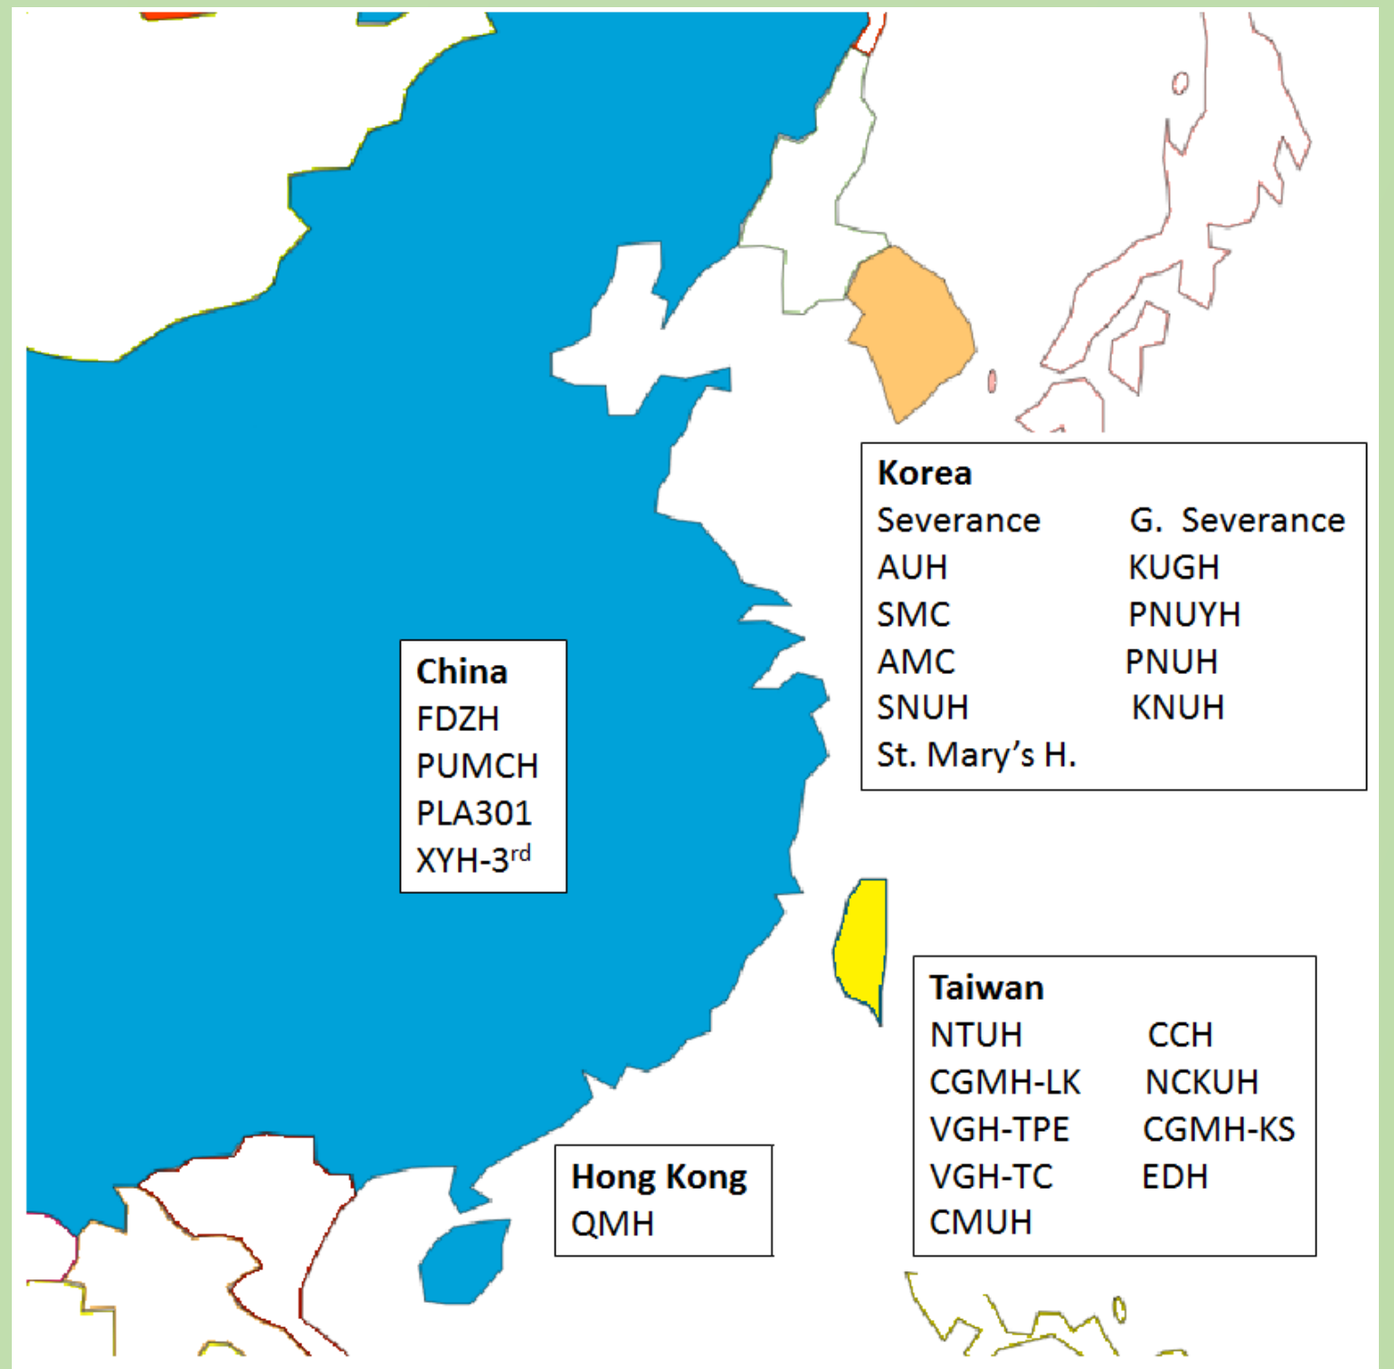

Korea
Severance
AUH
SMC
AMC
SNUH
St. Mary's H.
G. Severance
KUGH
PNUYH
PNUH
KNUH
China
FDZH
PUMCH
PLA301
XYH-3rd
Taiwan
NTUH
CGMH-LK
VGH-TPE
VGH-TC
CMUH
CCH
NCKUH
CGMH-KS
EDH
Hong Kong
QMH

Fig.3 Participating sites and regions for PATRON

Methods

This was a prospective, randomized, double-blind, placebo-controlled, parallel-group, international multicenter phase III trial of muparfostat conducted in the Asia-Pacific region (Taiwan, Korea, China, and Hong-Kong) with 25 sites and more than 140 investigators' participation from 2011 (Fig.3). A total of 520 HV-HCC patients after surgical resection were randomized (1:1) to receive injection of either muparfostat (160 mg/day, 4-days-on/3-days-off, 3-weeks-on/1-week-off) or placebo for 52 weeks and followed up for 96 weeks (Fig.4). The primary endpoint was centrally assessed disease-free survival (DFS). Secondary endpoints included overall survival (OS), time to recurrence, and safety.

Fig.4 PATRON study design and muparfostat administration regimen

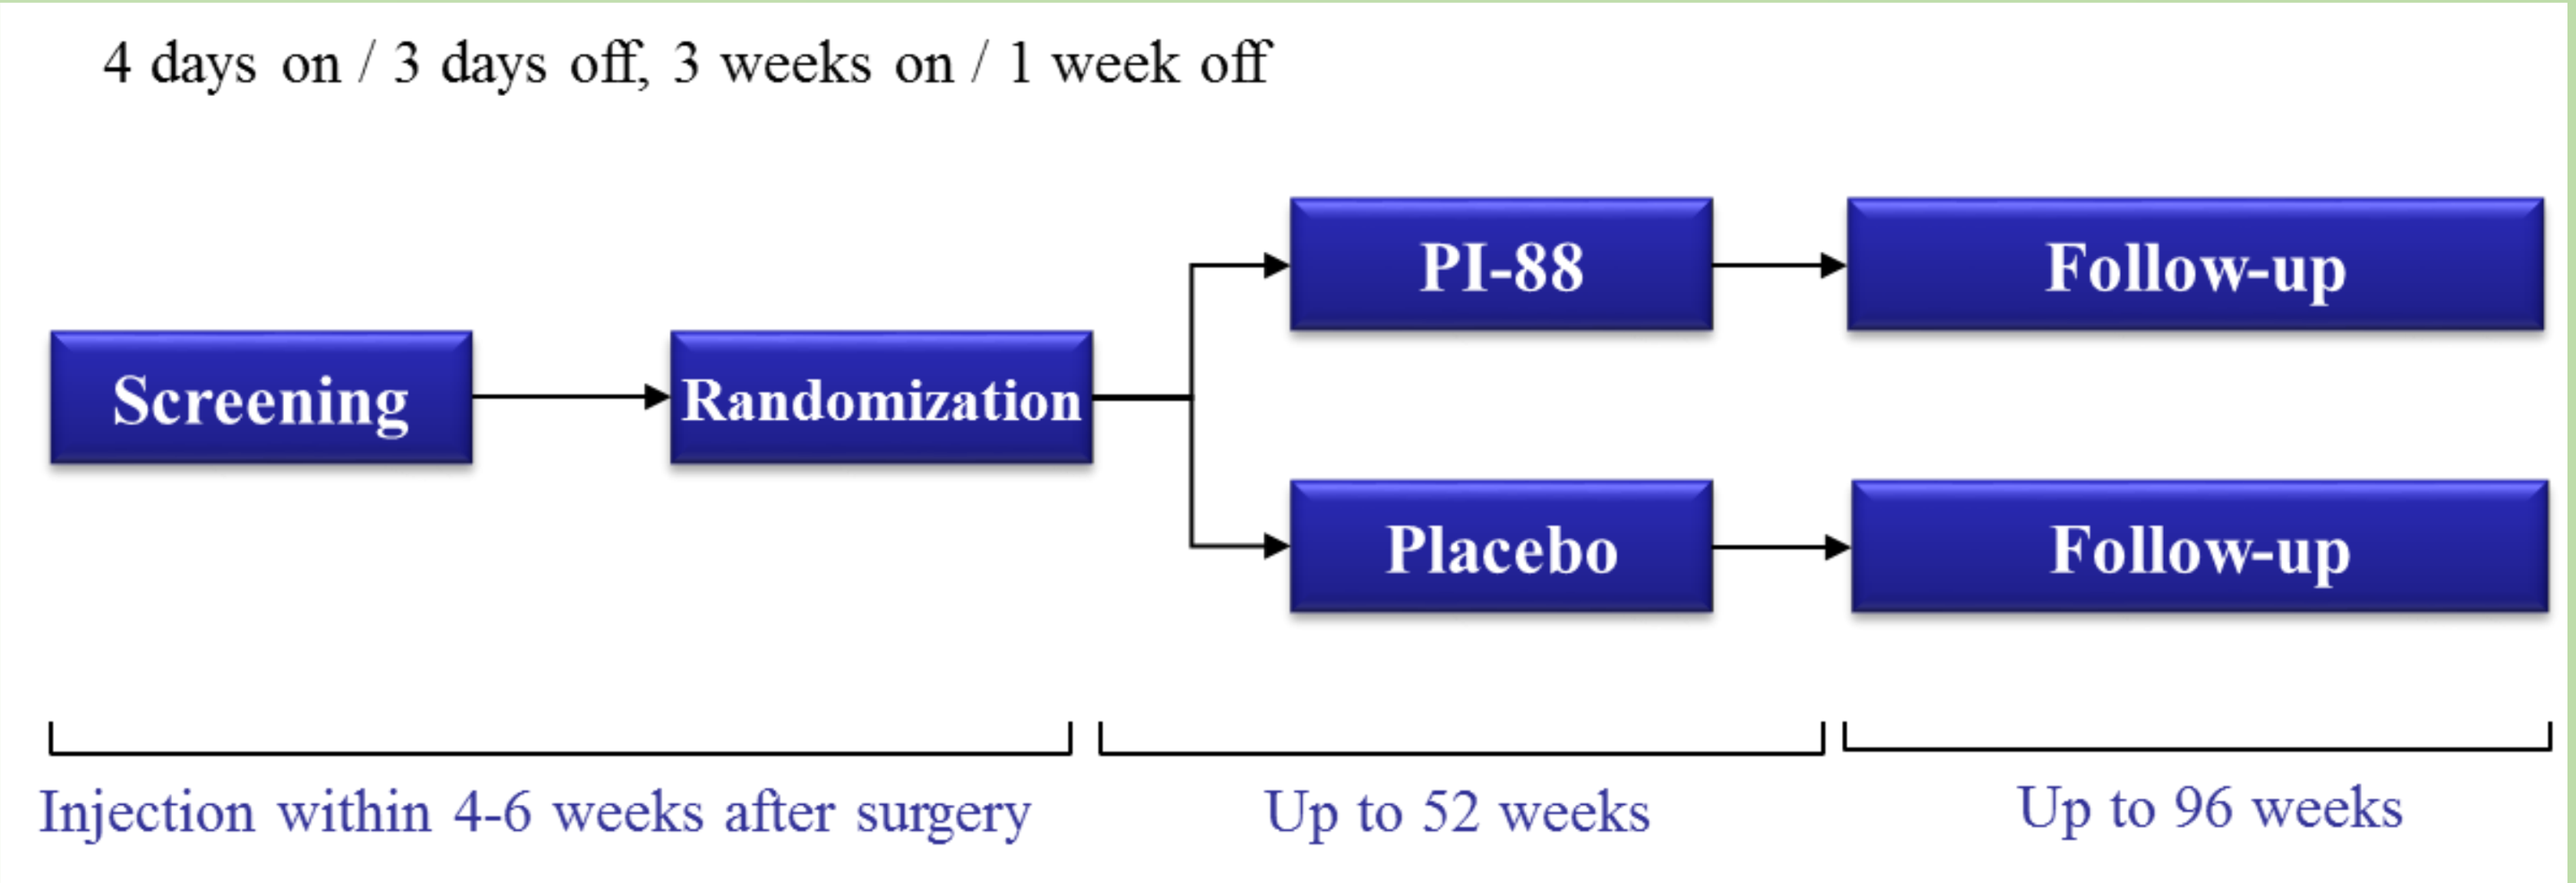

4 days on / 3 days off, 3 weeks on / 1 week off
Screening
Randomization
PI-88
Placebo
Follow-up
Follow-up
Injection within 4-6 weeks after surgery
Up to 52 weeks
Up to 96 weeks

Results

Baseline patient demographics and characteristics were balanced between the treatment and placebo arms (Table 1). After interim analysis in 2014, the trial was early concluded and data analyzed in 2017. The final intention-to-treat analysis (N = 519) yielded a non-significant result on DFS, not reaching the primary end point (Fig.5). However, per-protocol analysis (N = 423) revealed a positive protective effect in a distinct subgroup. Muparfostat showed benefits in patients with microvascular invasion on pathologic examination, which accounts for about 41% of all trial patients (Fig.6). If early recurrence within 24 weeks after treatment initiation was excluded from both arms, muparfostat group had a significant prolongation of the disease-free time after completion of the 1-year treatment in patients with microvascular invasion (hazard ratio: 0.2026, p = 0.0379) (Fig.7). Muparfostat had a good safety profile. There were five clinically suspected cases of heparin-induced thrombocytopenia but only one was confirmed. 30 (11.6%) subjects reported a total of 34 treatment emergent SAEs but only 2 possible drug-related in the treatment group, while 15 (5.8%) subjects with 15 treatment emergent SAEs in the placebo group, and no newly observed safety signals were raised in the study as compared with earlier trials.

Characteristics	Placebo	PI-88	Total	P-value
Age [years]				
N (Missing)	261 (0)	258 (0)	519 (0)	**0.2757**
Mean (SD)	55.08 (9.619)	54.12 (10.201)	54.61 (9.915)	
Sex				
N (Missing)	261 (0)	258 (0)	519 (0)	**0.3289**
Male	217 (83.1%)	206 (79.8%)	423 (81.5%)	
Female	44 (16.9%)	52 (20.2%)	96 (18.5%)	
ECOG Performance Score				
N (Missing)	261 (0)	258 (0)	519 (0)	**0.8495**
0	247 (94.6%)	245 (95.0%)	492 (94.8%)	
1	14 (5.4%)	13 (5.0%)	27 (5.2%)	
Child-Pugh Score (Post-OP)				
N (Missing)	261 (0)	258 (0)	519 (0)	**0.4220**
5	207 (79.3%)	198 (76.7%)	405 (78.0%)	
6	47 (18.0%)	53 (20.5%)	100 (19.3%)	
7	6 (2.3%)	3 (1.2%)	9 (1.7%)	
8	1 (0.4%)	4 (1.6%)	5 (1.0%)	
Number of Tumors				
N (Missing)	261 (0)	258 (0)	519 (0)	**0.2825**
1	235 (90.0%)	223 (86.4%)	458 (88.2%)	
2	20 (7.7%)	28 (10.9%)	48 (9.2%)	
≥3	6 (2.3%)	7 (2.7%)	13 (2.5%)	
Hepatitis Serology				
N (Missing)	260 (1)	258 (0)	518 (1)	**0.2306**
HBV+ & HCV+	13 (5.0%)	10 (3.9%)	23 (4.4%)	
HBV+ Only	222 (85.4%)	212 (82.2%)	434 (83.8%)	
HCV+ Only	20 (7.7%)	33 (12.8%)	53 (10.2%)	
HBV- & HCV-	5 (1.9%)	3 (1.2%)	8 (1.5%)	

Table 1 Baseline patient demographics (abbreviated)

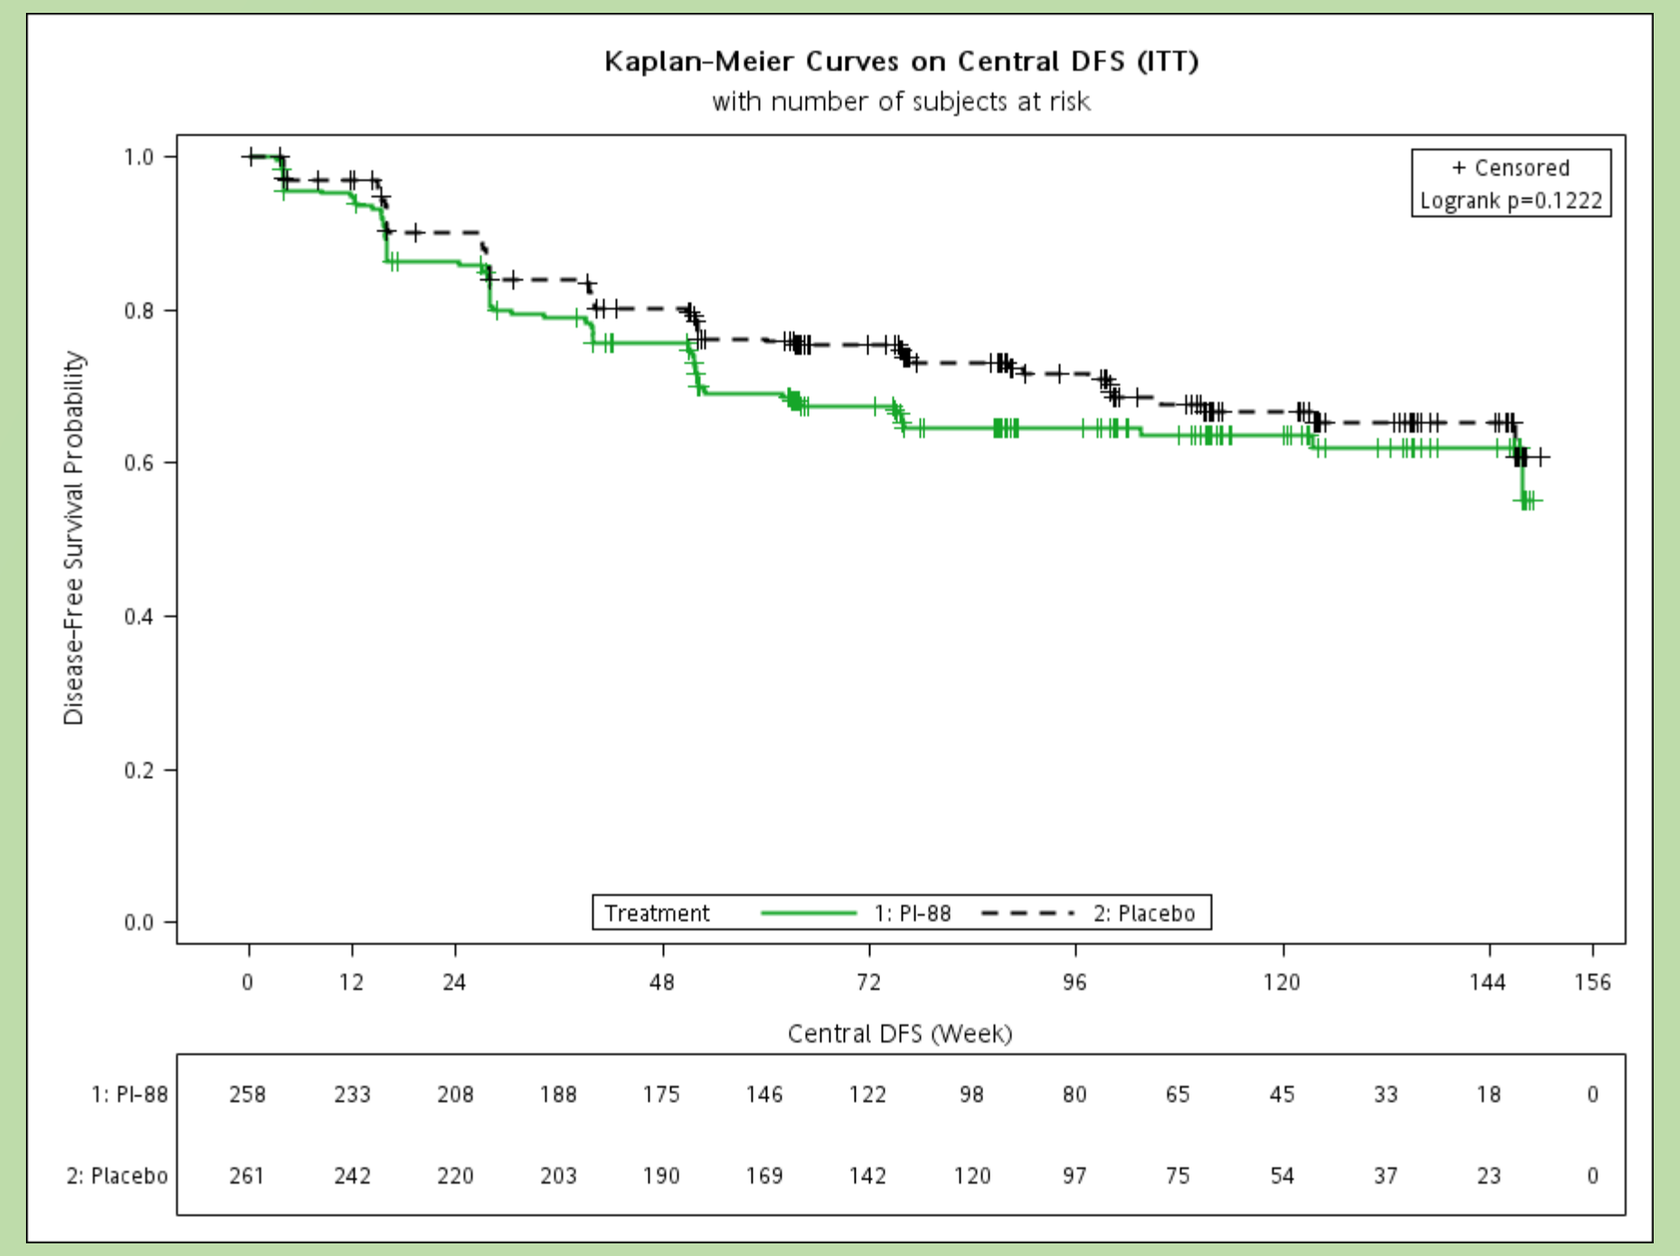

Kaplan-Meier Curves on Central DFS (ITT)
with number of subjects at risk
+ Censored
Logrank p=0.1222
Disease-Free Survival Probability
Treatment 1: PI-88 2: Placebo
Central DFS (Week)
1: PI-88 258 233 208 188 175 146 122 98 80 65 45 33 18 0
2: Placebo 261 242 220 203 190 169 142 120 97 75 54 37 23 0

Fig.5 Results of primary endpoint analysis in ITT population

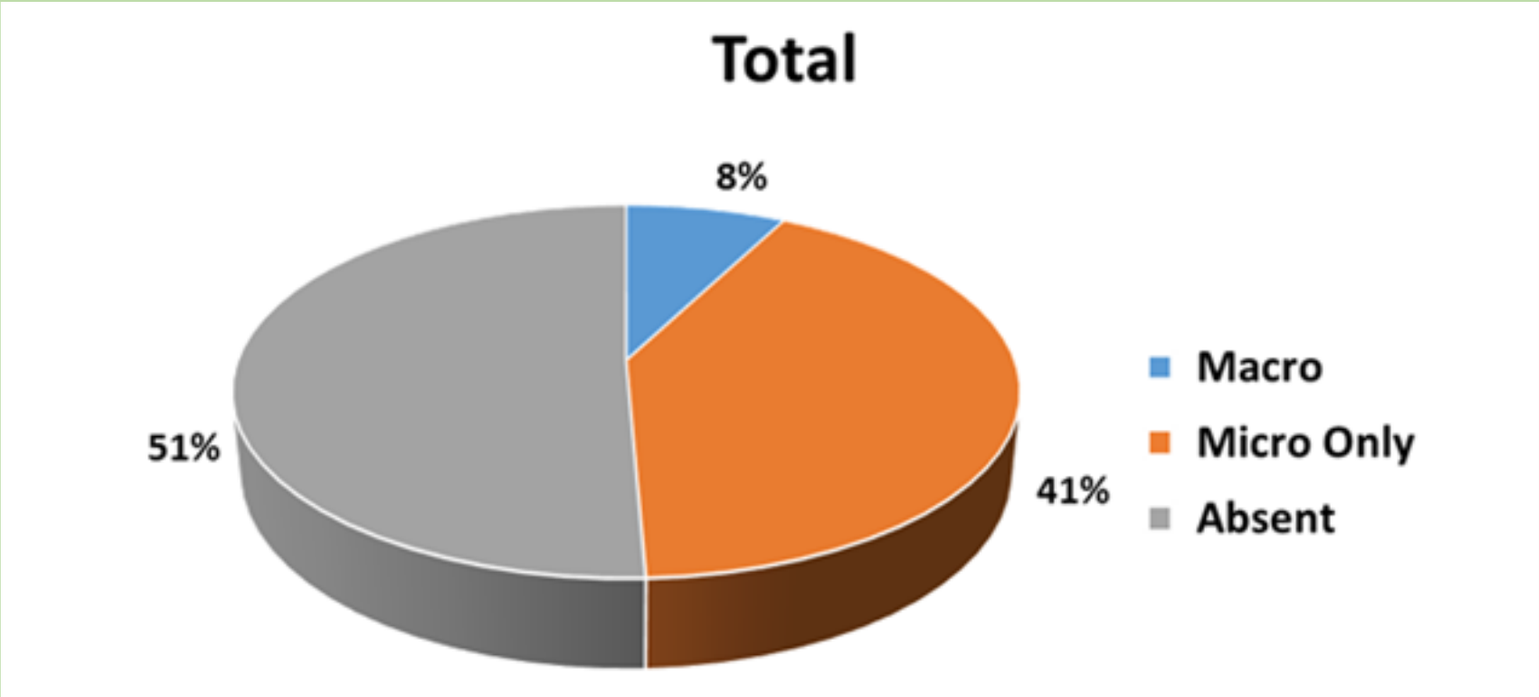

Total
8%
51%
41%
Macro
Micro Only
Absent

Group	Placebo	PI-88	Total
N	261	258	519
Macro	22 (8.4%)	18 (7.0%)	40 (7.7%)
Micro	**105 (40.2%)**	**106 (41.1%)**	**211 (40.7%)**
Absent	134 (51.3%)	134 (51.9%)	268 (51.6%)

Fig.6 HCC tumor vascular invasion subtype distributions in PATRON study

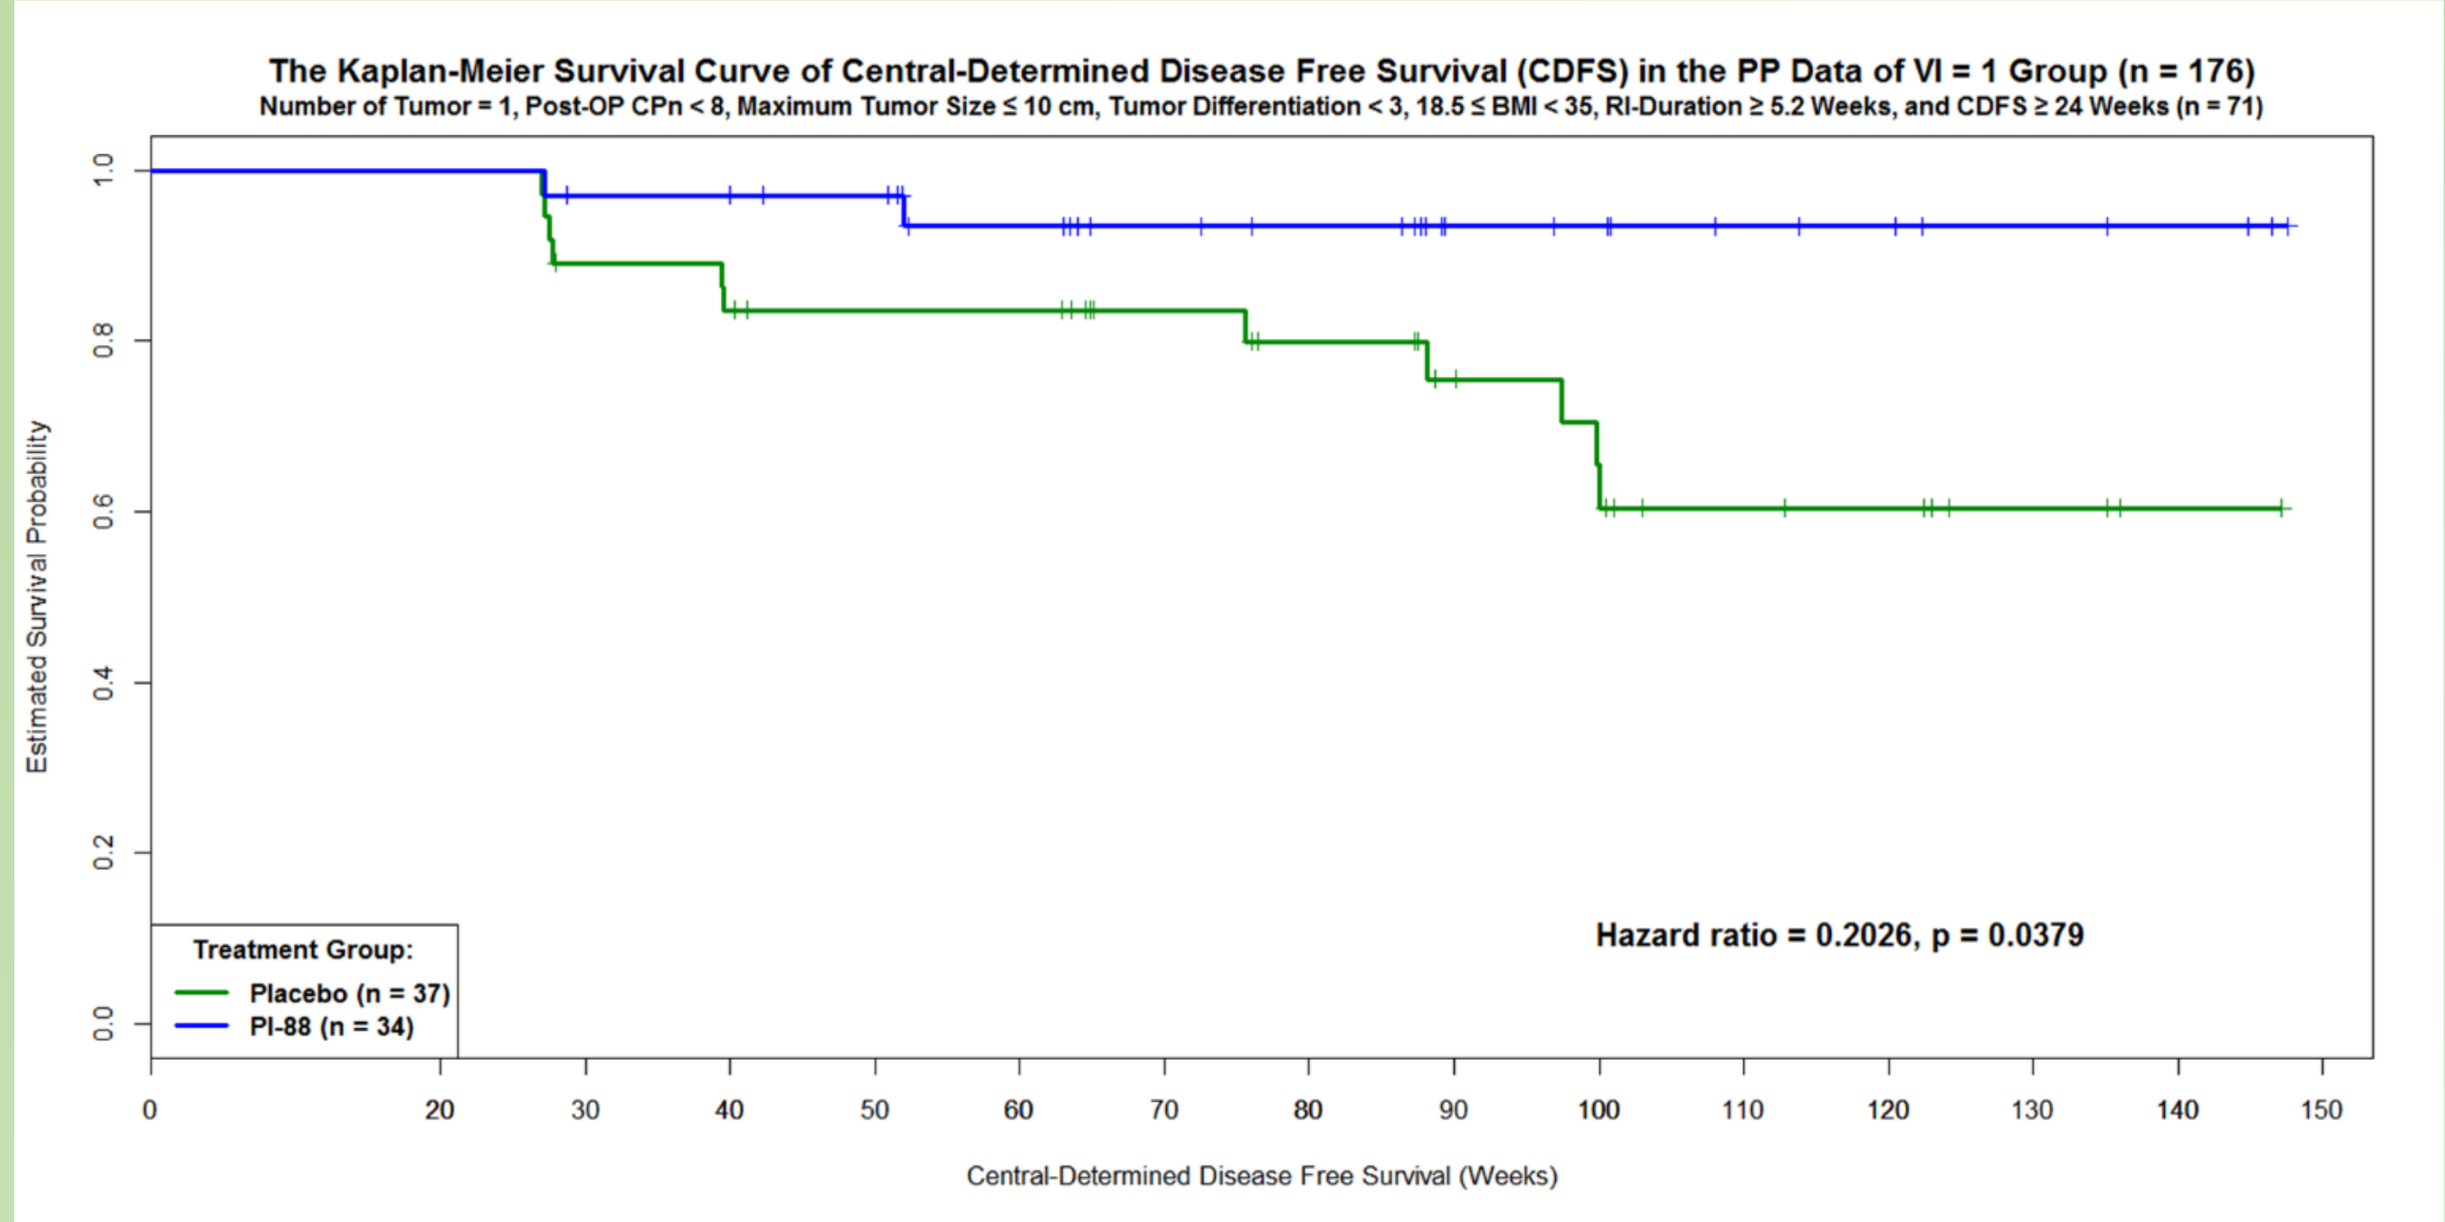

The Kaplan-Meier Survival Curve of Central-Determined Disease Free Survival (CDFS) in the PP Data of VI = 1 Group (n = 176)
Number of Tumor = 1, Post-OP CPn < 8, Maximum Tumor Size ≤ 10 cm, Tumor Differentiation < 3, 18.5 ≤ BMI < 35, RI-Duration ≥ 5.2 Weeks, and CDFS ≥ 24 Weeks (n = 71)
Estimated Survival Probability
Hazard ratio = 0.2026, p = 0.0379
Treatment Group:
Placebo (n = 37)
PI-88 (n = 34)
Central-Determined Disease Free Survival (Weeks)

Fig.7 Per-protocol subgroup analysis by central reading

Conclusions

Despite the fact that DFS was not improved in the overall treatment group, muparfostat could significantly prolong the DFS in the microvascular-invasion subgroup, comprising 41% of the trial population. With recent growing knowledge of heparanase and other growth factors that play roles in tumor microenvironment modulations, our study results suggest the potential of muparfostat as a single therapy or in combination with other anti-cancer agents for future HCC adjuvant therapy trials.

References

1. Vlodavsky, et al. Cancer Microenvironment. 2012
2. Heyman, et al. Experimental Hematology. 2016
3. Liu, et al. J Hepatol. 2009.
4. Liu, et al. World J Gastroenterol. 2014.

Corresponding author: PJ Chen
peijerchen@ntu.edu.tw

This study has received funding from:
Medigen Biotechnology Corp. and
Taiwan MOEA Technology Development Program